Reused with permission from the American Society of Hematology. © 2024/2023 The Authors. All rights reserved. Officially licensed by ASH for distribution via https://www.adctmedical.com/ and reactively by ADC Therapeutics.

A phase II trial of loncastuximab tesirine in patients with previously treated Waldenström macroglobulinemia

Jorge J. Castillo; Steve P. Treon; Catherine A Flynn; Andres Ramirez-Gamero; Nina Budano; Alexandra N. Eurell; Kirsten Meid; Shayna Sarosiek
Bing Center for Waldenström Macroglobulinemia, Dana-Farber Cancer Institute; Department of Medicine, Harvard Medical School

Background

- Chemoimmunotherapy and covalent BTK inhibitors are standard therapies often used in first and second-line therapy for Waldenström macroglobulinemia (WM).
- These therapies are well tolerated, but there is no standard of care for patients who progress on both of these treatments.
- This prospective phase II clinical trial was designed to evaluate the use of loncastuximab tesirine, a CD19 antibody drug conjugate, in patients with WM previously treated with at least two lines of therapy, including rituximab and a BTK inhibitor.

Methods

- This multicenter, phase II trial is enrolling previously treated symptomatic patients with WM.
- Loncastuximab tesirine as a single agent is given once every 4 weeks for a total of 6 doses.
- Loncastuximab is administered in cycles 1-2 at 150 µg/kg and cycles 3-6 at 75 µg/kg.
- Baseline testing includes complete blood counts, chemistry panel, and IgM levels as well as a bone marrow biopsy/aspirate with molecular testing and PET or CT imaging of the chest/abdomen/pelvis.
- The primary outcome measure is overall response rate (ORR).
- Secondary outcomes are evaluating the depth of hematologic response, the median progression free survival, the effect of bone marrow disease burden on disease response, the impact of MYD88 and CXCR4 on disease response, the safety of loncastuximab tesirine in WM, the rate of IgM flare, the rate of tumor lysis, and patient quality of life during treatment.
- Assumptions included H0 ≤35%, H1 ≥65%, 2-sided alpha 0.035, and power 0.85. The trial is designed to enroll 36 patients.
- For purposes of this interim analysis, >2 of the first 7 participants would have to attain a response to study therapy to reject futility.

Results

- At the time of data cut-off 10 patients have enrolled in the trial and 8 patients have initiated treatment
- No IgM flare and no tumor lysis syndrome have occurred

Baseline Characteristics (n = 8)

	N (range or %)
Median age, y	68 (53-77)
Male sex	4 (50%)
Median previous therapies	4 (2-6)
Median serum IgM, mg/dL	1967 (723-5955)
Median hemoglobin, g/dL	9.7 (8.9-12.6)
Median bone marrow involvement, %	65 (43-90)
MYD88 MUT	8 (100%)
CXCR4 MUT	7 (88%)
TP53 MUT	5 (63%)
Lymphadenopathy ≥1.5 cm	1 (13%)
Splenomegaly ≥15 cm	1 (13%)

Hematologic responses (n = 8)

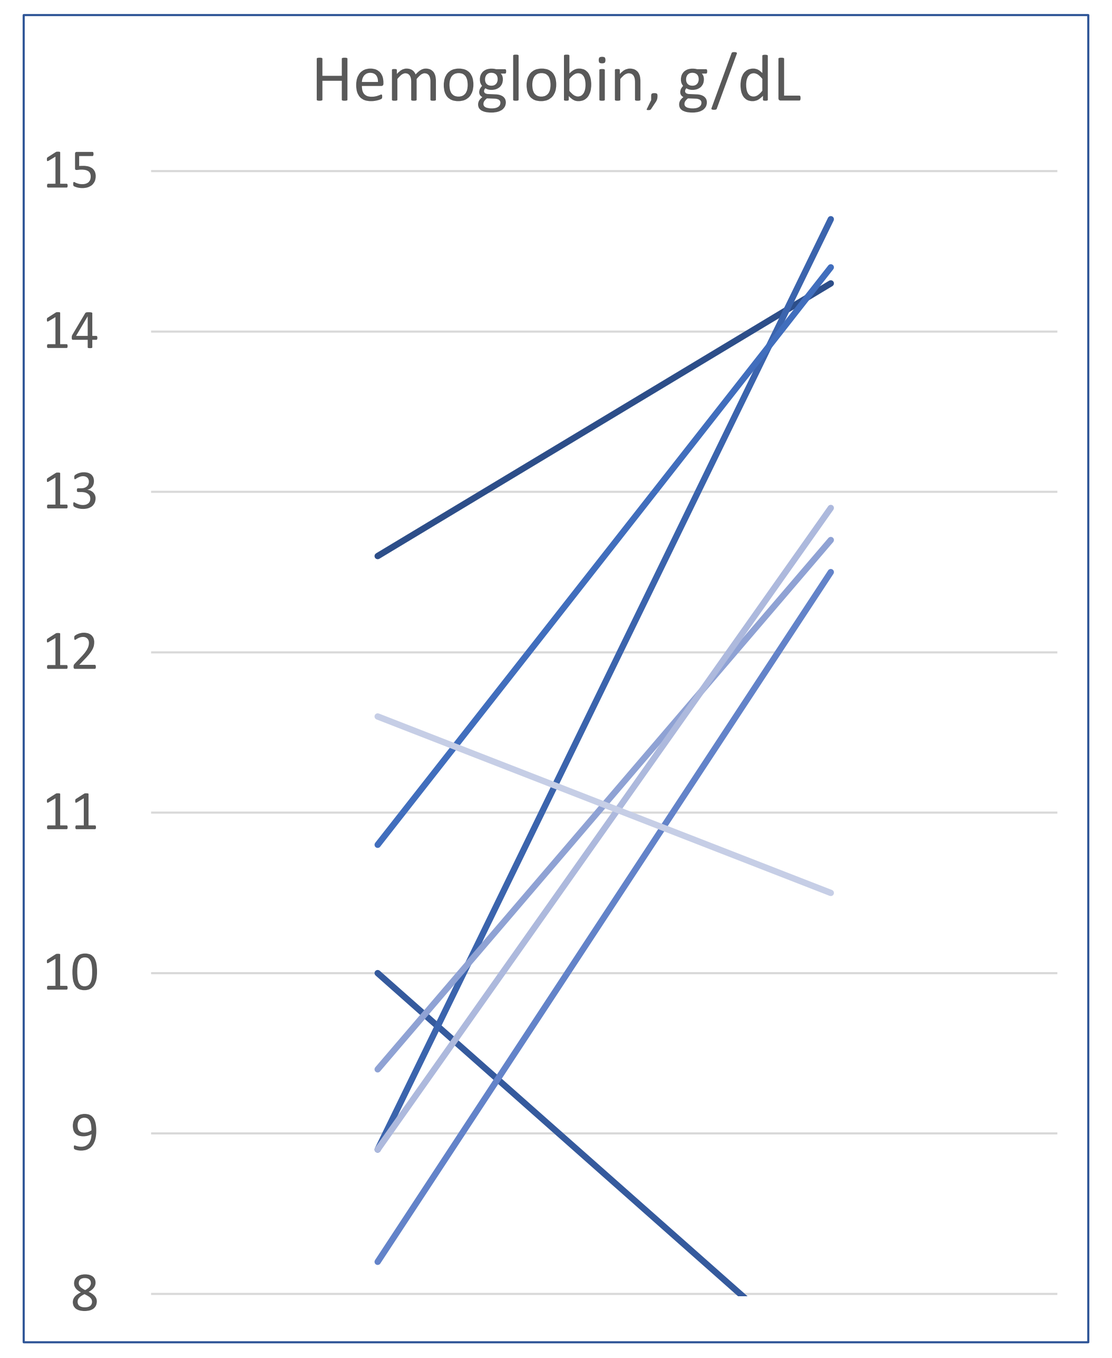
Hemoglobin, g/dL
15
14
13
12
11
10
9
8

Median post-treatment hgb: 12.9 g/dL

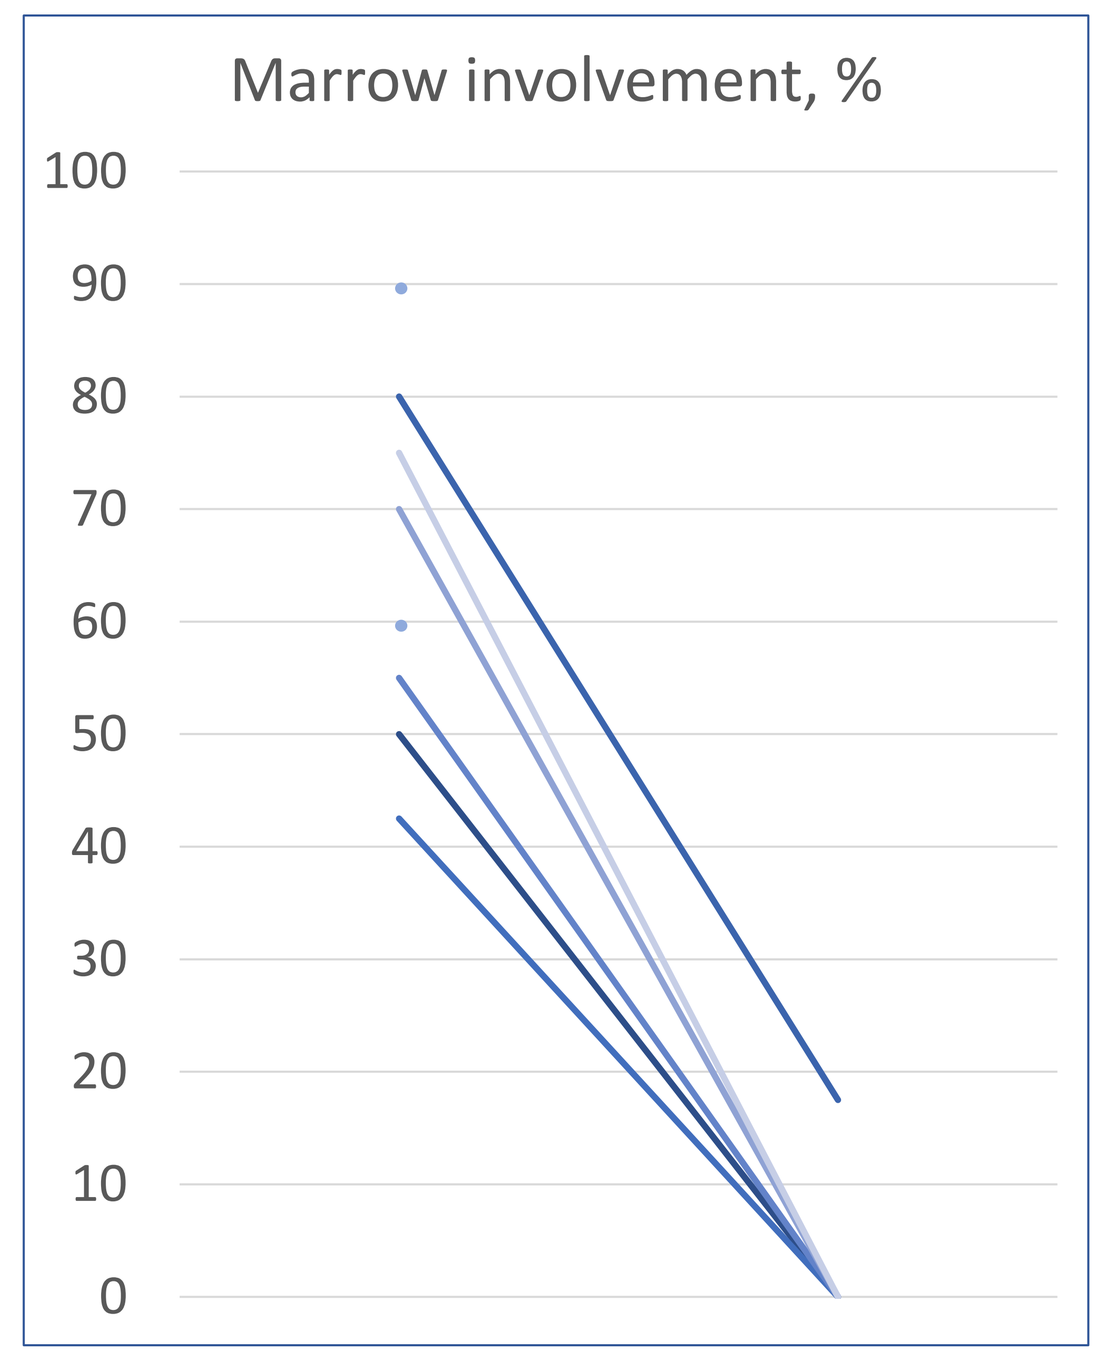
Marrow involvement, %
100
90
80
70
60
50
40
30
20
10
0

Median post-treatment marrow: <5%

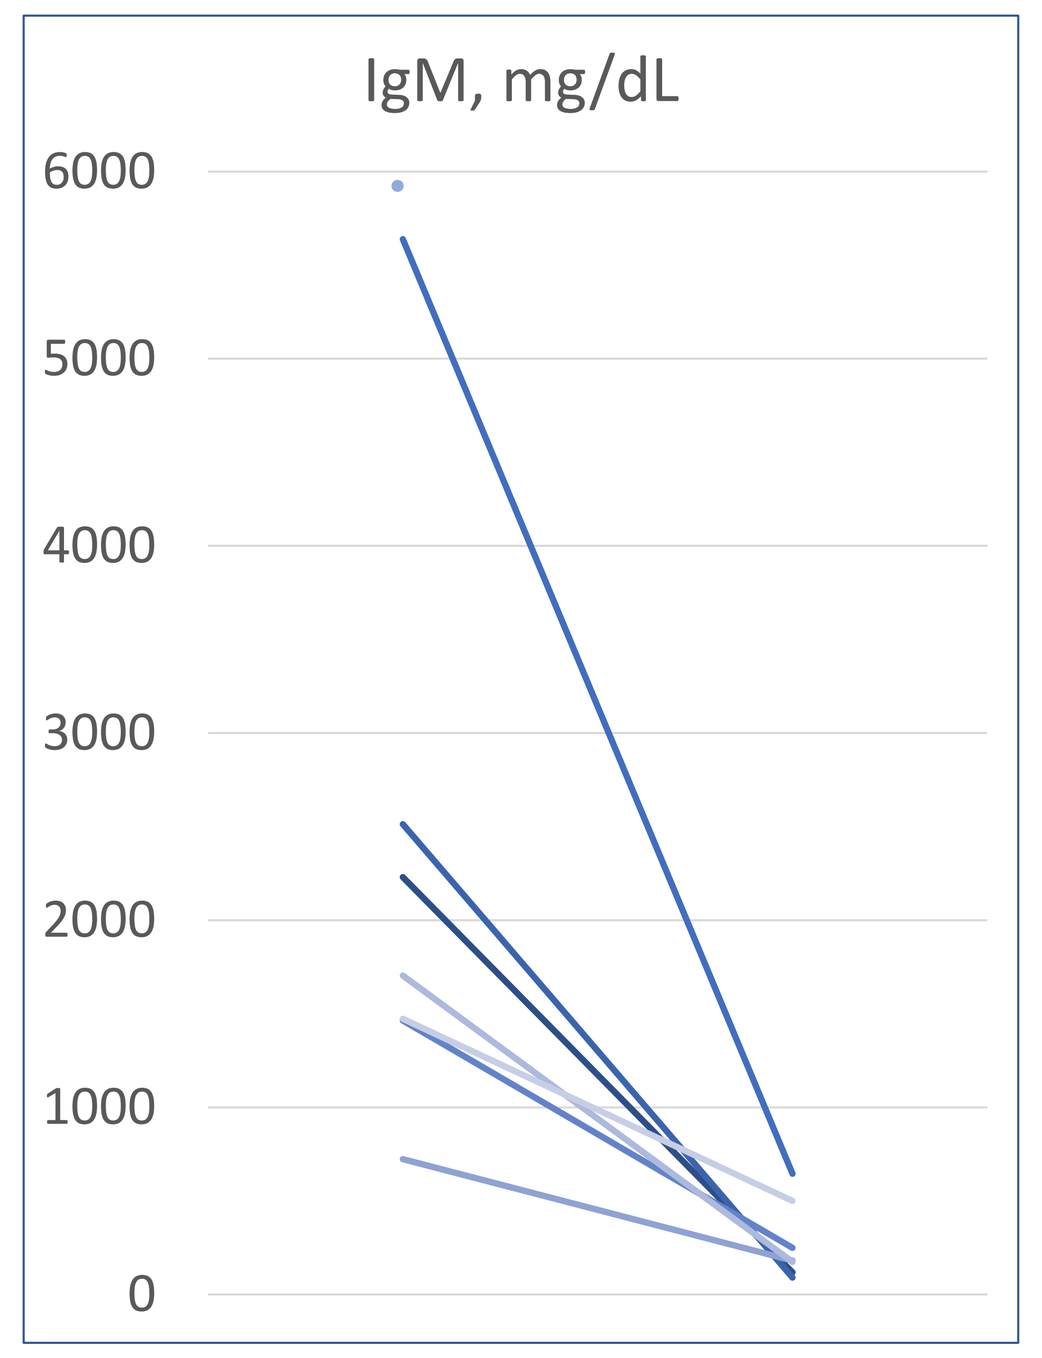
IgM, mg/dL
6000
5000
4000
3000
2000
1000
0

Median post-treatment IgM: 181 mg/dL

*2 patients did not have a post-treatment bone marrow biopsy and 1 patient was removed from the study due to disease progression after 1 cycle with ongoing pheresis and therefore did not have an evaluable post-treatment IgM (these patients are indicated by a single dot on the above plots)

Hematologic responses (n=8)

- Overall response rate: 88%
- Major response rate: 88%
- Median time to minor response: 13 days (range 4-77)
- Median time to major response: 54 days (range 14-105)

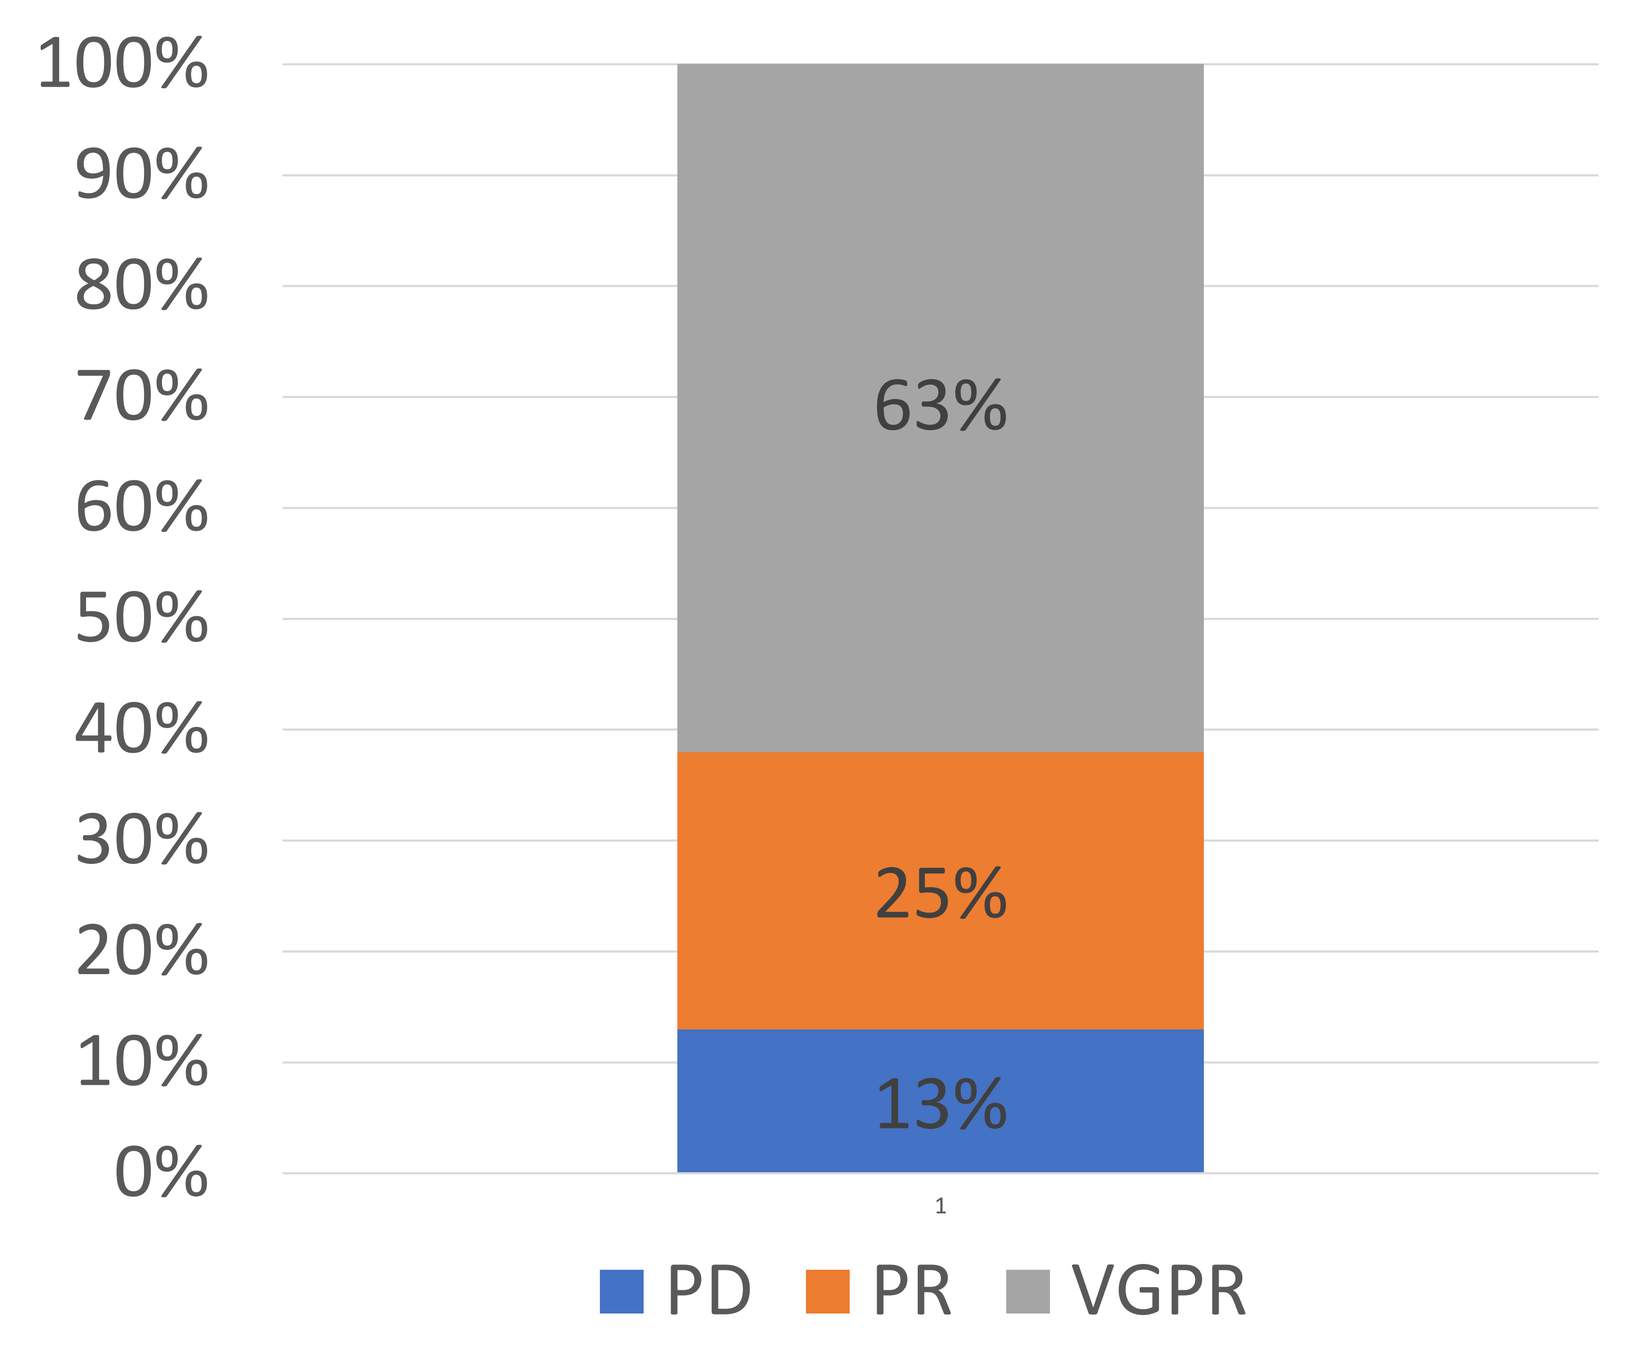
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
63%
25%
13%
1
PD
PR
VGPR

Toxicity

- Toxicities are similar to that seen in prior loncastuximab trials, with skin toxicity or rash in 88% (grades 1-2), edema in 63% (grades 1-3), cytopenias in 100% (grades 1- 4), and asymptomatic GGT elevation in 63% (grades 1-4).
- 3/8 patients received only 1 cycle due to disease progression (n=1), grade 4 GGT elevation (n=1), and patient decision (n=1).
- 3/8 patients required dose reduction due to hematologic toxicity.
- Although skin toxicities and GGT elevation have persisted in some patients post-treatment, cytopenias have been transient.

Conclusions

- Early data from this ongoing clinical trial demonstrates an overall response rate of 88% in the initial 8 patients treated with this fixed duration therapy.
- This ORR compares favorably to other third line agents in WM.
- This treatment has a manageable side effect profile with skin toxicity, asymptomatic GGT elevation, and transient cytopenias.
- Accrual will continue to enroll 36 total patients.

Contact Information

- Shayna_sarosiek@dfci.harvard.edu